Poster 177

Treatment Outcomes With Unselected Autologous Tumor-Infiltrating Lymphocytes in Patients With Checkpoint Inhibition–Refractory Advanced Cutaneous Melanoma

Robert E. Hawkins,[1-3] Yizhou Jiang,[2] Paul C. Lorigan,[1] Fiona C. Thistlethwaite,[1,3] Manon Pillai,[2] Martine Thomas,[2] Natalia Kirillova,[2] John S. Bridgeman,[2] Gray Kueberuwa,[2] Ryan D. Guest,[2] and Zachary J. Roberts[2]

[1]The Christie, NHS Foundation Trust, Manchester, United Kingdom; [2]Instil Bio, Inc, Dallas, TX, USA; and [3]University of Manchester, Manchester, United Kingdom

InstilBio

BACKGROUND

- Most patients with advanced melanoma who receive programmed cell death protein 1 inhibitor (PD-1i) therapy develop treatment resistance; those who relapse after PD-1i (and if, *B-Raf proto-oncogene, serine/threonine kinase [BRAF]*- and/or *mitogen-activated protein kinase kinase [MEK]*-mutated, BRAF and/or MEK inhibition) have poor outcomes and limited treatment options[1-3]
- The intrinsic antitumor activity and unrestricted T-cell receptor repertoire of unselected autologous tumor-infiltrating lymphocytes (TILs) may provide advantages over other treatments in solid tumors, including checkpoint inhibitor therapy-refractory melanoma.[1,4,5] TIL therapy has shown durable complete responses (CRs) in patients with melanoma, with an estimated 41% objective response rate in advanced cutaneous melanoma[6]
- In a retrospective analysis of a single-center compassionate use clinical series of 21 patients with advanced melanoma, TIL products made from tumor digests showed a high overall response rate (ORR; 67%) and CR rate (19%) and a safety profile consistent with that of lymphodepletion and high-dose interleukin (IL)-2[7]
- This subanalysis of the compassionate use clinical series assesses outcomes for patients who received TILs after prior PD-1 inhibition, a patient subset with limited treatment options

METHODS

PATIENTS

Table 1. Guidelines to Determine Suitability of Patients to Receive TILs Within This Clinical Series

Should Have	Should Not Have
• Histologically confirmed malignant melanoma with confirmed evidence of progressive metastatic disease • No standard-of-care treatment options • Satisfactory hematologic and biochemical indices • Adequate cardiac function • Suitable fitness for all planned treatments and procedures (including surgery for TIL harvest, lymphodepleting chemotherapy, TILs, and IL-2) • A metastatic site that could be excised to obtain a specimen of at least 1 cm³; for lymph nodes, these must have been >2 cm³ • Measurable/evaluable disease after the surgical resection	• Prior allogeneic transplantation • Symptomatic brain metastasis measuring ≥10 mm in diameter • Lymphotoxic therapy such as chemotherapy, high-dose steroids, or other immunosuppressive therapy within 4 weeks of harvesting • Concurrent serious infection within 28 days prior to treatment • Steroid use ≤3 weeks before treatment, except for physiological replacement doses of steroids

IL-2, interleukin-2; TIL, tumor-infiltrating lymphocyte.

TREATMENT

Figure 1. Tissue Procurement and Manufacturing

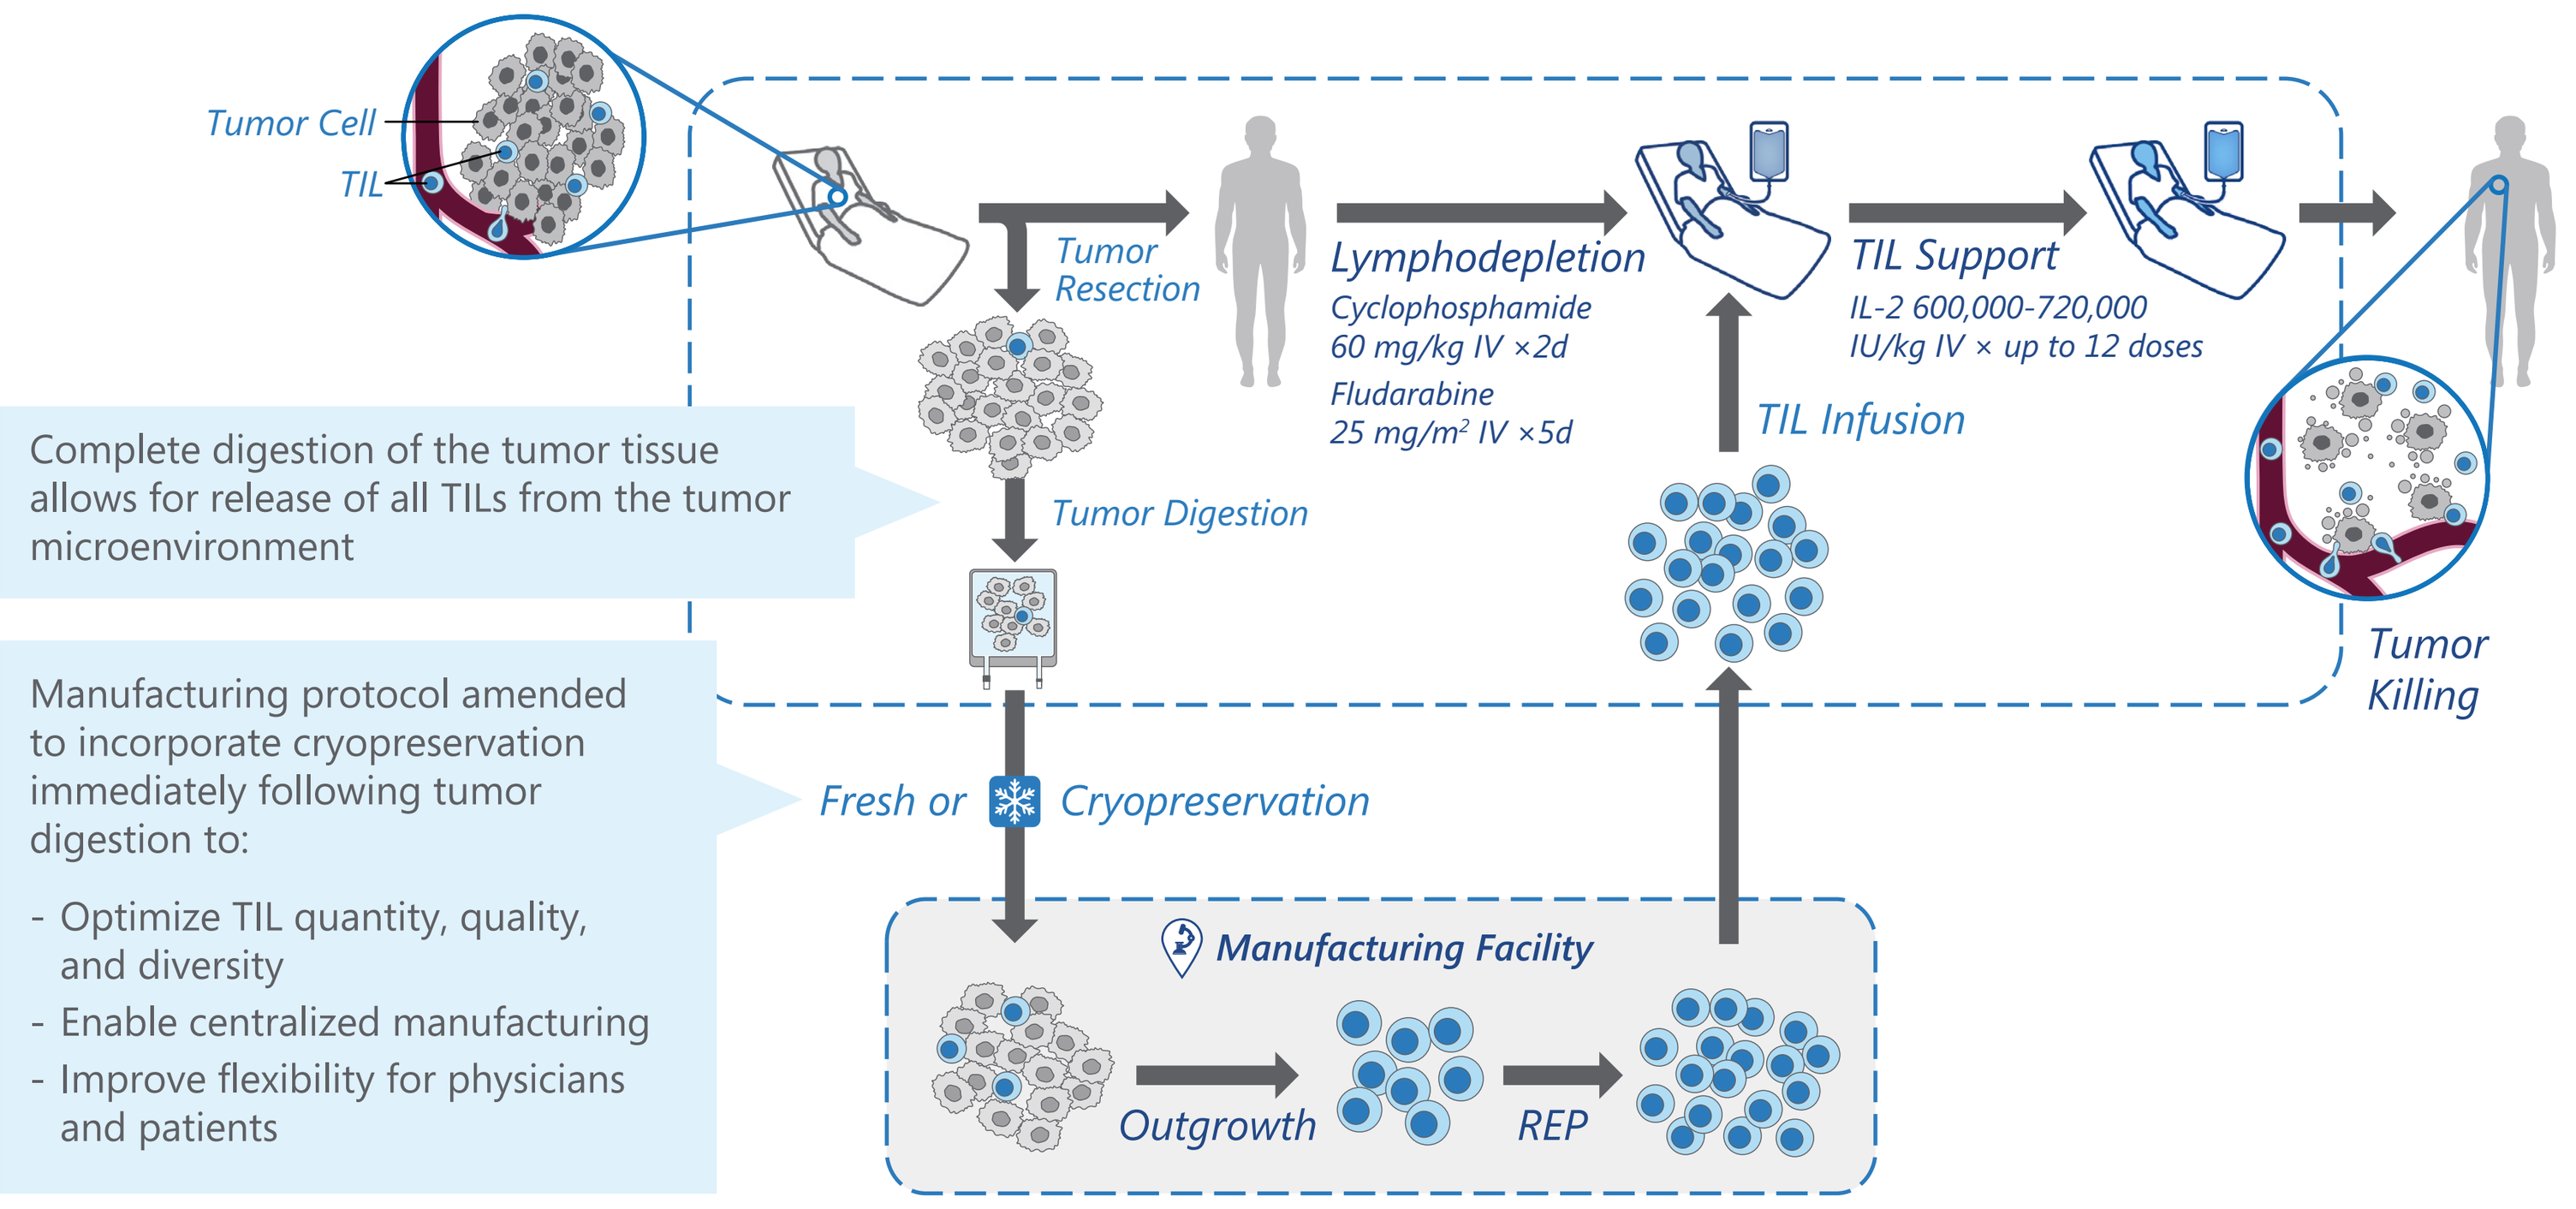
Tumor Cell
TIL
Tumor Resection
Lymphodepletion
Cyclophosphamide 60 mg/kg IV ×2d
Fludarabine 25 mg/m² IV ×5d
TIL Support
IL-2 600,000-720,000 IU/kg IV × up to 12 doses
TIL Infusion
Tumor Digestion
Tumor Killing
Complete digestion of the tumor tissue allows for release of all TILs from the tumor microenvironment
Manufacturing protocol amended to incorporate cryopreservation immediately following tumor digestion to:
- Optimize TIL quantity, quality, and diversity
- Enable centralized manufacturing
- Improve flexibility for physicians and patients
Fresh or
Cryopreservation
Manufacturing Facility
Outgrowth
REP

IL-2, interleukin-2; IV, intravenous; REP, rapid expansion protocol; TIL, tumor-infiltrating lymphocyte.

- Unselected autologous TILs derived from digested tumor tissue were manufactured under a Medicines and Healthcare Products Regulatory Agency Manufacturing Specials license
- Patients received lymphodepleting chemotherapy (cyclophosphamide 60 mg/kg/d ×2 days, fludarabine 25 mg/m^2/d ×5 days) followed by TIL infusion and post-TIL short course of high-dose IL-2 (600,000-720,000 IU/kg) on a compassionate use basis
- Patients were hospitalized for treatment

METHODS *(CONTINUED)*

ASSESSMENTS AND ANALYSES

- Patients who received prior PD-1i therapy were included in the subanalysis
- Safety was assessed by clinically significant adverse events (AEs) with onset post-TIL infusion, as reported during the hospitalization period
- Efficacy for all patients was assessed locally with pre- and post-treatment imaging
 - Nine of 12 patients underwent posttreatment imaging and quantitative tumor volume assessments consistent with Response Evaluation Criteria in Solid Tumors version 1.1 (RECIST 1.1)
 - The remaining 3 patients were followed with imaging, including positron emission tomography, computed tomography, and magnetic resonance imaging as well as clinical monitoring (eg, history and physical examination, laboratory assessments), but did not have quantitative tumor volume measurements
- Data cutoff date was December 31, 2019

RESULTS

Table 2. Treatment Exposure

Treatment Exposure	Prior PD-1i Subgroup (n=12)
Received lymphodepleting chemotherapy, n (%)	12 (100)
Received TIL treatment, n (%)	12 (100)
Total TIL cells infused (×10^9), median (range)	32.4 (7.9-53.0)
No. of IL-2 doses, median (range)	8 (6-9)

IL-2, interleukin-2; PD-1i, programmed cell death protein 1 inhibitor; TIL, tumor-infiltrating lymphocyte.

- Between October 2011 and August 2019, 21 patients with advanced cutaneous melanoma were treated
- Of 21 patients, 12 received prior PD-1i therapy and are reported herein
- Median follow-up time for the 12 PD-1i–treated patients was 45.5 months

Table 3. Demographics and Baseline Characteristics

	Prior PD-1i Subgroup (n=12)	All Treated Patients (N=21)
Age, median (range), y	55 (33-64)	45 (16-68)
Male, n (%)	7 (58)	15 (71)
Stage IV, n (%)	12 (100)	21 (100)
Disease sites, median (range)	4 (2–10)	4 (2–10)
M1c disease, n (%)	9 (75)	14 (67)
M1d disease, n (%)	3 (25)	7 (33)
Tumor burden,[a] median (range), mm	123 (51-169)[b]	100 (13-281)[b]
Lactate dehydrogenase level, n (%)		
>ULN to ≤2×ULN	4 (33)	7 (33)
>2×ULN	2 (17)	3 (14)
No. of prior systemic regimens, mean (range)	3 (1-9)	3 (1-9)
Checkpoint inhibitor, n (%)	12 (100)	19 (91)
PD-1i	12 (100)	12 (57)
CTLA-4i	12 (100)	19 (91)
Dual PD-1i/CTLA-4i relapsed/refractory	12 (100)	12 (57)
BRAF-mutated patients, n (%)	6 (50)	11 (52)
BRAFi±MEKi	6 (50)	11 (52)

BRAF, B-raf proto-oncogene, serine/threonine kinase; BRAFi, BRAF inhibitor; CTLA-4, cytotoxic T-lymphocyte–associated protein 4; CTLA-4i, CTLA-4 inhibitor; MEK, mitogen-activated protein kinase kinase; MEKi, MEK inhibitor; PD-1, programmed cell death protein 1; PD-1i, PD-1 inhibitor; RECIST, Response Evaluation Criteria in Solid Tumors; ULN, upper limit of normal.
[a]Target lesions sum of diameters (local assessment per RECIST 1.1).
[b]Tumor burden data at baseline were available for all but 1 patient.

- Patients had highly advanced disease with high baseline tumor burden and were heavily pretreated
- All patients were dual relapsed/refractory to PD-1i/cytotoxic T-lymphocyte–associated protein 4 inhibitor (CTLA-4i) therapy, and all *BRAF*-mutated patients received prior BRAF inhibitor (BRAFi) therapy alone or in combination with MEK inhibitor (MEKi) therapy

RESULTS *(CONTINUED)*

Table 4. Summary of Safety

Any-Grade TEAEs Post-TIL Infusion ≥20%, n (%)	Prior PD-1i Subgroup (n=12)
Thrombocytopenia	9 (75)
Pyrexia	6 (50)
Rigors	6 (50)
Vascular leak	4 (33)
Chest infection	3 (25)
Neutropenia	3 (25)

TEAE, treatment-emergent adverse event; TIL, tumor-infiltrating lymphocyte.

- Safety profile of the prior PD-1i subgroup was consistent with that of the all-treated population[7] and published literature[6]
- AEs were managed supportively and mostly attributable to lymphodepleting chemotherapy and IL-2
- No treatment-related deaths occurred
- Six patients died >3 months after TIL infusion and before the data cutoff date
 - Three due to progressive disease (PD)
 - One possibly due to adverse event caused by subsequent anticancer therapy
 - Two with documented PD before death, but specific cause of death was not available

Table 5. Best Overall Response

Response	Prior PD-1i Subgroup (n=12)	All Treated Patients[a] (N=21)
Best overall response (CR+PR), n (%)	7 (58)	14 (67)
CR	1 (8)	4 (19)
PR	6 (50)	10 (48)
SD, n (%)	2 (17)	4 (19)
PD, n (%)	3 (25)	3 (14)
Disease control rate (CR+PR+SD), n (%)	9 (75)	18 (86)
Median time to response, mo	1.8	1.7

CR, complete response; PD, progressive disease; PR, partial response; SD, stable disease; TIL, tumor-infiltrating lymphocyte.
[a]Responders include 2 patients (1 in the prior PD-1i subgroup) with dabrafenib plus mitogen-activated protein kinase kinase inhibitor-refractory disease whose disease was unequivocally progressing on the combination therapy before TIL and who received postinfusion dabrafenib to prevent tumor flare.

- Responses were generally consistent between patients who received prior PD-1i therapy and all treated patients
- Responses were similar among RECIST-evaluable patients (n=9/12) and all patients who received prior PD-1i
 - ORR in the RECIST-evaluable prior PD-1i subgroup was 56%, including 1 patient (11%) with CR

Figure 2. Overall Survival

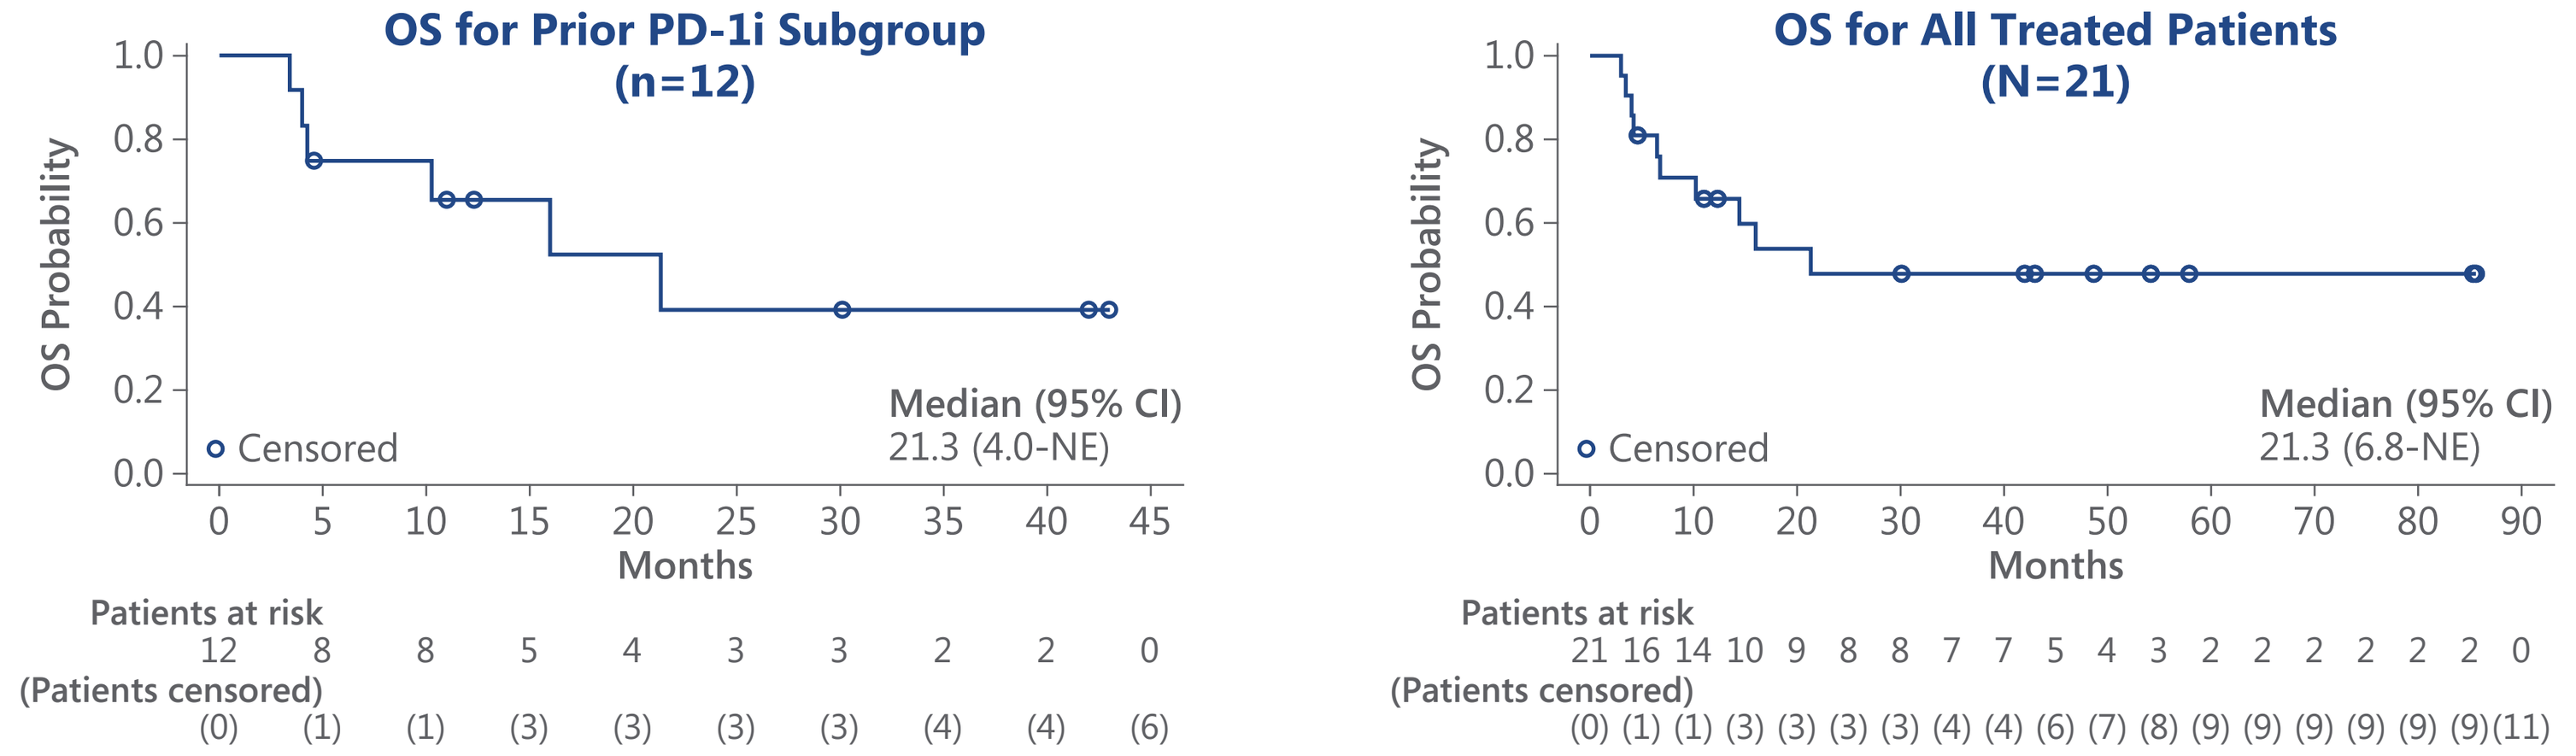
OS for Prior PD-1i Subgroup (n=12)
OS Probability
1.0
0.8
0.6
0.4
0.2
0.0
Censored
Median (95% CI)
21.3 (4.0-NE)
0 5 10 15 20 25 30 35 40 45
Months
Patients at risk
12 8 8 5 4 3 3 2 2 0
(Patients censored)
(0) (1) (1) (3) (3) (3) (3) (4) (4) (6)
OS for All Treated Patients (N=21)
OS Probability
Censored
Median (95% CI)
21.3 (6.8-NE)
0 10 20 30 40 50 60 70 80 90
Months
Patients at risk
21 16 14 10 9 8 8 7 7 5 4 3 2 2 2 2 2 2 0
(Patients censored)
(0) (1) (1) (3) (3) (3) (3) (4) (4) (6) (7) (8) (9) (9) (9) (9) (9) (9)(11)

NE, not evaluable; OS, overall survival; PD-1i, programmed cell death protein 1.

- Median OS in patients who received prior PD-1i therapy and in the all treated patient population was 21.3 months

Figure 3. Time to Response in Responding Patients (n=7)

Patient	Prior Checkpoint Inhibitor	RECIST-Evaluable[a]
1	Ipi and pembro	Yes
2	Ipi and nivo	Yes
3[b]	Ipi and nivo	No
4	Ipi and pembro	Yes
5[c]	Ipi and nivo	No
6	Ipi and pembro	Yes
7	Ipi, nivo, and pembro	Yes

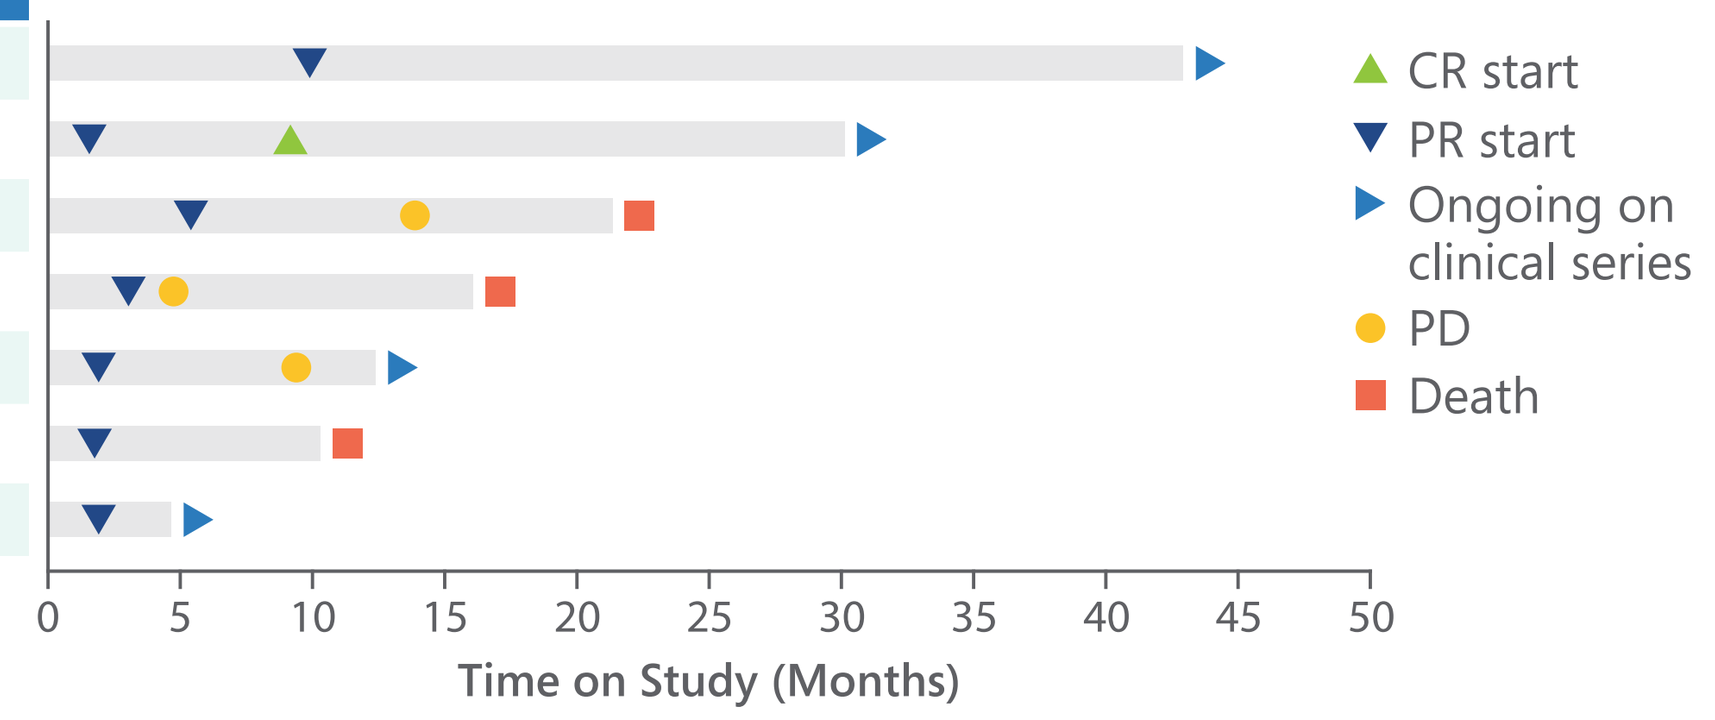
CR start
PR start
Ongoing on clinical series
PD
Death
0 5 10 15 20 25 30 35 40 45 50
Time on Study (Months)

CR, complete response; IL-2, interleukin-2; ipi, ipilimumab; nivo, nivolumab; PD, progressive disease; pembro, pembrolizumab; PR, partial response; RECIST; Response Evaluation Criteria in Solid Tumors; SD, stable disease; TIL, tumor-infiltrating lymphocyte.
[a]Four additional RECIST-evaluable patients are not depicted as these patients were nonresponders.
[b]Patient 3 had unequivocally B-raf proto-oncogene inhibitor– and mitogen-activated protein kinase kinase inhibitor–refractory melanoma immediately before TIL treatment but was continued on dabrafenib, with brief interruptions for tumor harvest and TIL infusion, to prevent tumor flare on discontinuation.
[c]Patient 5 received checkpoint inhibitor before documented disease progression.

- With a median follow-up of 45.5 months, 2 of 12 patients (17%) had durable ongoing responses (>30 months post-TIL infusion)

Figure 4. Case Study: Tumor Resection for TIL Harvest Before PD-1i Therapy Followed by Post–PD-1i TIL Administration

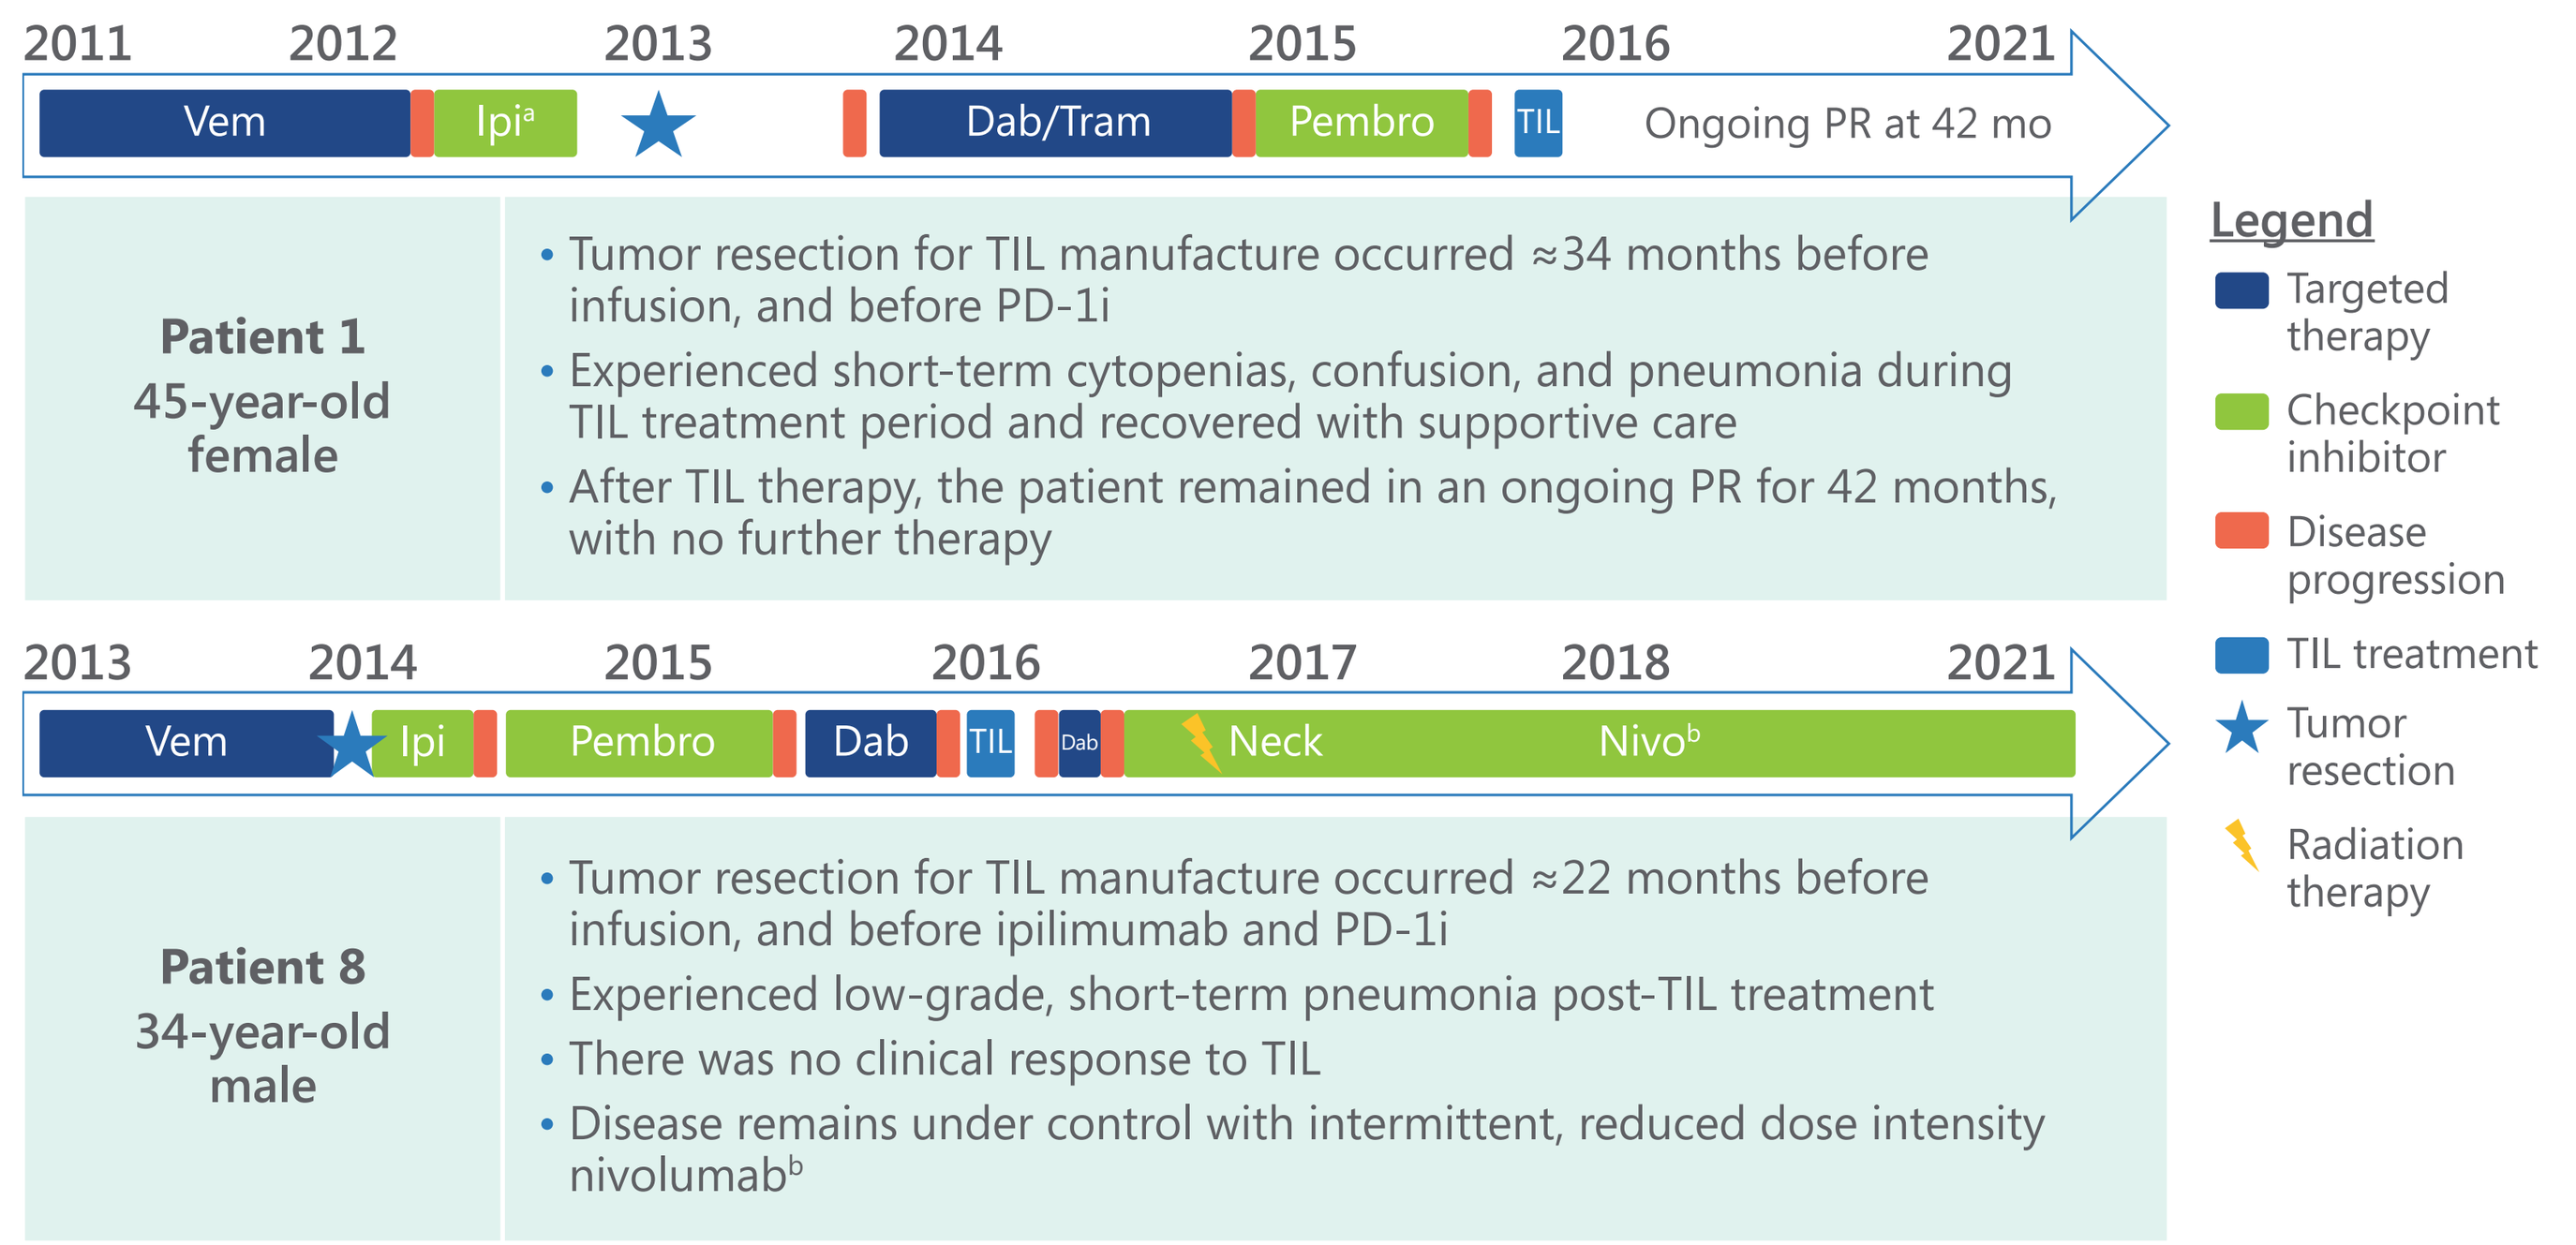
2011 2012 2013 2014 2015 2016 2021
Vem
Ipi[a]
Dab/Tram
Pembro
TIL
Ongoing PR at 42 mo
Patient 1
45-year-old female
• Tumor resection for TIL manufacture occurred ≈34 months before infusion, and before PD-1i
• Experienced short-term cytopenias, confusion, and pneumonia during TIL treatment period and recovered with supportive care
• After TIL therapy, the patient remained in an ongoing PR for 42 months, with no further therapy
2013 2014 2015 2016 2017 2018 2021
Vem
Ipi
Pembro
Dab
TIL
Dab
Neck
Nivo[b]
Patient 8
34-year-old male
• Tumor resection for TIL manufacture occurred ≈22 months before infusion, and before ipilimumab and PD-1i
• Experienced low-grade, short-term pneumonia post-TIL treatment
• There was no clinical response to TIL
• Disease remains under control with intermittent, reduced dose intensity nivolumab[b]
Legend
Targeted therapy
Checkpoint inhibitor
Disease progression
TIL treatment
Tumor resection
Radiation therapy

AE, adverse event; Dab, dabrafenib; Ipi, ipilimumab; Nivo, nivolumab; PD-1i, programmed cell death protein 1 inhibitor; Pembro, pembrolizumab; TIL, tumor-infiltrating lymphocyte; Tram, trametinib; Vem, vemurafenib.
[a]Patient presented with hypophysitis while on ipi, requiring hormone replacement with glucocorticoids; no pembro-associated toxicity was observed.
[b]Patient presented with colitis/pneumonitis while on nivo, which was managed by reducing nivo frequency (given every 6 weeks); patient remains in stable disease with reduced nivo dosing, and the aforementioned toxicity is no longer evident.

- In 2 patients with poor-risk disease who had tumor resection for TIL harvest before PD-1i treatment, TILs remained active despite intercurrent PD-1 inhibition
- After TIL treatment, there was no recurrence of immune-related AEs in either patient

CONCLUSIONS

- In this subanalysis of patients with relapsed advanced melanoma after both PD-1i and CTLA-4i, and for some, BRAFi, outcomes of unselected autologous TILs were similar to those observed in all treated patients, with high response rates and a safety profile consistent with that of TIL therapy[6]
- Unselected TILs may address the unmet medical need for the poor-risk subset of patients with advanced melanoma who experience disease progression after checkpoint inhibition and, if applicable, targeted therapy
- Two patients underwent tumor harvest well before PD-1i therapy and then TIL infusion after progression on checkpoint and BRAF/MEK inhibitor(s), suggesting that early tumor collection and banking may offer additional options for patients with early or high-risk disease
 - One patient achieved durable disease control to PD-1i retreatment after progression on TIL therapy, suggesting sensitivity to PD-1i may be restored by TIL therapy
- These retrospective results are limited by small sample size, and further studies are warranted
- DELTA-1, a global phase 2 clinical trial of this therapy in patients with advanced melanoma, is currently enrolling patients (NCT05050006; EudraCT 2020-003862-37)

REFERENCES

1. van den Berg JH, et al. *J Immunother Cancer.* 2020;8:e000848.
2. Schadendorf D, et al. *Lancet.* 2018;392:971-984.
3. Weichenthal M, et al. *J Clin Oncol.* 2019;37:9505.
4. Sarnaik A, et al. *J Clin Oncol.* 2021;39:2656-2666.
5. Rosenberg SA, et al. *Clin Cancer Res.* 2011;17:4550-4557.
6. Dafni U, et al. *Ann Oncol.* 2019;30:1902-1913.
7. Hawkins RE, et al. *Cancer Res.* 2021;81(13):LB150.

ACKNOWLEDGMENTS

- We would like to thank all the staff within The Christie NHS Foundation Trust and The Christie Clinic who worked tirelessly to provide high-quality care to all the patients in this report
- Medical writing support was provided by Christopher Waldapfel, PharmD, of Instil Bio, Inc, and Jennifer Yang, PhD, of Nexus GG Science with funding from Instil Bio, Inc

DISCLOSURES

REH: employment with and stock or other ownership in Instil Bio, Inc; consultancy or advisory role for Anaveon AG, Novalgene, Ltd, Servier, Celyad, Celgene, Oxford Biomedica, GSK, Bristol Myers Squibb, Gilead, EUSA Pharma, Novartis, and Pfizer. **YJ:** employment with and stock or other ownership in Instil Bio, Inc. **PCL:** consulting or advisory role for Amgen, Bristol Myers Squibb, MSD, Novartis, Nektar, and PierreFabre; speakers' bureau participation for Bristol Myers Squibb, MSD, Novartis, and PierreFabre; research funding from Bristol Myers Squibb; and travel support from Bristol Myers Squibb and MSD. **FCT:** consulting or advisory role for Achilles, Bayer, Bristol Myers Squibb, Evelo Therapeutics, GSK, T-knife, and Zelluna Immunotherapy; research funding from Novartis; and coordinating or local physician investigator for AbbVie, Achilles Ltd, Adaptimmune, Agalimmune, AstraZeneca, AVEO, Bristol Myers Squibb, Chugai Pharmaceutical Co, CytomX, Daiichi Sankyo, GenMab, GSK, Immunocore, Incyte, Janssen, Kymab Ltd, Millenium Pharmaceuticals/Takeda, Novartis, Pfizer, Roche, and Synthon. **MP:** speakers' bureau participation for Bristol Myers Squibb, Ipsen, Pfizer, and Novartis; travel support from EUSA Pharma and Bristol Myers Squibb. **MT:** employment with Cellular Therapeutics, Ltd, Immetacyte, Ltd, and Instil Bio, Inc. **NK:** former employment with and stock or other ownership in Immetacyte, Inc, and stock or other ownership in Instil Bio, Inc. **JSB:** employment with and stock or other ownership in Cellular Therapeutics, Immetacyte, Inc, and Instil Bio, Inc. **GK:** employment with and stock or other ownership in Instil Bio, Inc. **RDG:** employment with and stock or other ownership in Instil Bio, Inc, Cellular Therapeutics, Ltd, and Immetacyte, Ltd, and named on associated patent applications owned by Instil Bio, Inc. **ZJR:** employment with and stock or other ownership in Instil Bio, Inc.

Copies of this e-Poster obtained through QR code are for personal use only and may not be reproduced without written permission of the authors.

Presented at the Society for Immunotherapy of Cancer (SITC) Annual Meeting, November 10-14, 2021.